BION-1301: a Novel fully Blocking APRIL Antibody for the Treatment of IgA Nephropathy

John Dulos[1], Wout Janssen[1], Lilian Driessen[1], Astrid Bertens[1], David Lutje Hulsik[1], Paul Vink[1], Hans van Eenennaam[1] and Andrea van Elsas[1]
[1]Aduro Biotech Europe, Oss, The Netherlands

INTRODUCTION

BION-1301 is a first-in-class humanized antibody targeting a proliferation Inducing Ligand (APRIL, TNFSF13). APRIL is a ligand for the receptors BCMA and TACI. APRIL serum levels are elevated in patients diagnosed with IgA Nephropathy (IgAN) and treatment with an anti-mouse APRIL antibody reduced serum IgA and proteinuria increase in an IgAN mouse model (Kim et al. PLOS One 2015). Altogether the data suggest that the APRIL-axis is important in IgAN pathology and strongly supports the development of Aduro's anti-APRIL antibody, BION-1301, to treat IgA Nephropathy (IgAN). BION-1301 is currently in a dose-escalation trial in Multiple Myeloma patients (NCT03340883).

Fig.1 BION-1301 Unique Epitope Mapped to Receptor Binding Sites

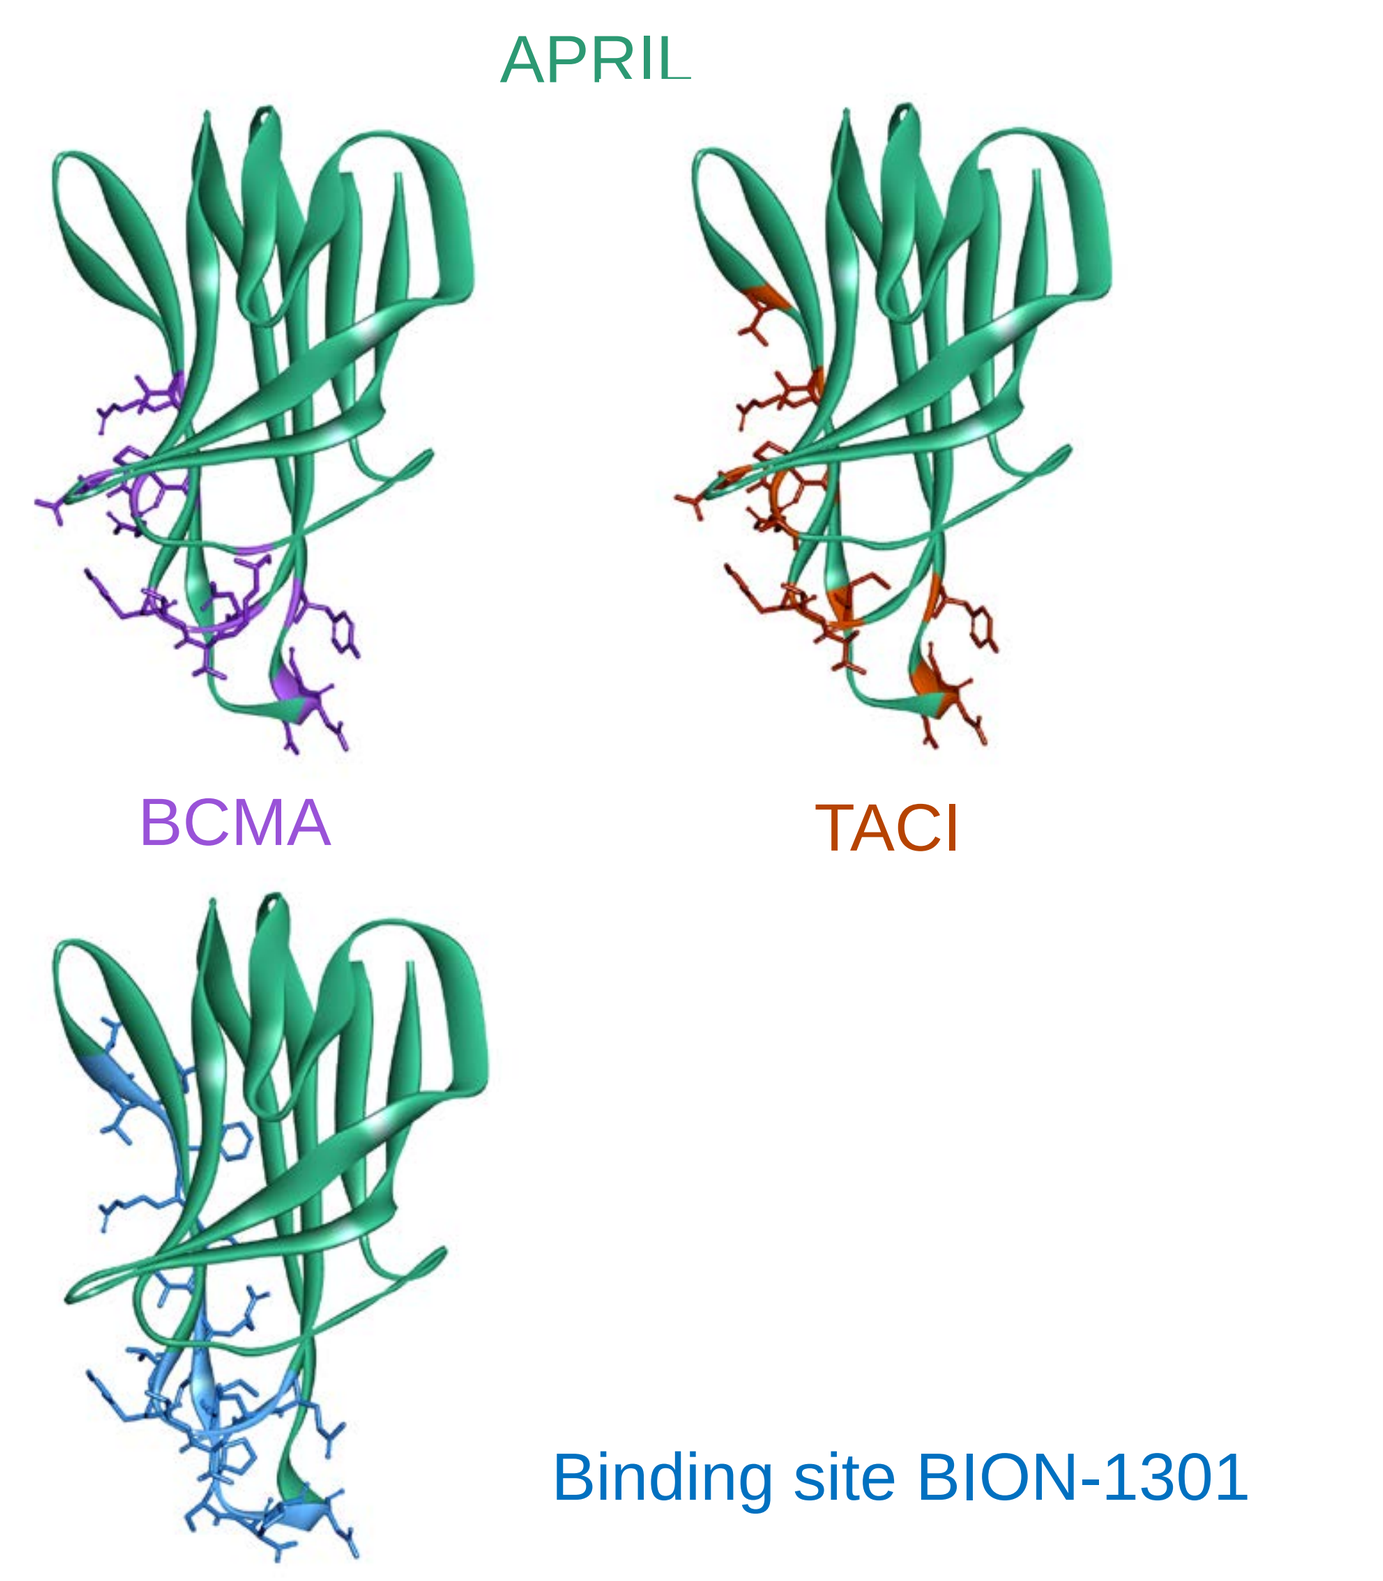
APRIL
BCMA
TACI
Binding site BION-1301

BION-1301 (blue) shares its APRILbinding site with that of both receptors TACI (purple) and BCMA (orange). Full blocking capacity to BION-1301 (Dulos et al. Blood 2016).

Fig.3 BION-1301 Single and Multiple Dose Studies in Non-Human Primates (NHP); No BION-1301 Related Toxicity

A)

Group	Treatment	No. of Females	Dose Level (mg/kg)
1	Control	3	0
2	BION-1301	3	0.3
3	BION-1301	3	3
4	BION-1301	3	30
5	Control	3	0
6	BION-1301 and TNP Ficoll	3	0.3
7	BION-1301 and TNP Ficoll	3	3
8	BION-1301 and TNP Ficoll	3	30

Single dose PK/PD study in NHP (A)
BION-1301 was dosed as a single dose via IV injection to female cynomolgus primates. Animals in Groups 1 to 4 (Phase 1 - PK) were observed post-dose for 84 days, whereas those in Groups 5 to 8 (Phase 2 - PK/PD) were observed post-dose for 42 days to assess PK, tolerability, reversibility, persistence or delayed occurrence of potential adverse pharmacology. TNP-Ficoll immunization was included in Groups 5 to 8 to assess PK/PD relationship of BION-1301 dose and exposure on T cell-independent B-cell responses. Results are presented in Fig. 4 and 6.

B)

Group	Treatment	No. of Animals		Dose Level (mg/kg)
		Male	Female	
1	Control	5	5	0
2	BION-1301	5	5	10
3	BION-1301	5	5	30
4	BION-1301	5	5	100

GLP toxicity multiple dose study in NHP (B)
BION-1301 was dosed IV weekly at 10, 30 or 100 mg/kg for 4 weeks (5 doses). On Day 31 of the dosing phase, terminal necropsy was performed on 3 subjects of each sex/group. Following a 12-week recovery period remaining animals underwent necropsy. Assessment of toxicity was based on mortality, clinical observations, body weights, food consumption, ophthalmic observations, electrocardiographic (ECG) measurements, physical examinations, pulse oximetry, blood pressure measurements, neurological examinations, dermal observations, clinical and anatomic pathology. Blood samples were collected for PK, PD, and immunogenicity evaluations. There were no BION-1301-related changes in organ weights, macroscopic findings or microscopic findings in animals sacrificed at the terminal necropsy.

Fig.2 BION-1301 Blocks APRIL-Induced B-Cell Proliferation and IgA Production *in vitro* and *in vivo*

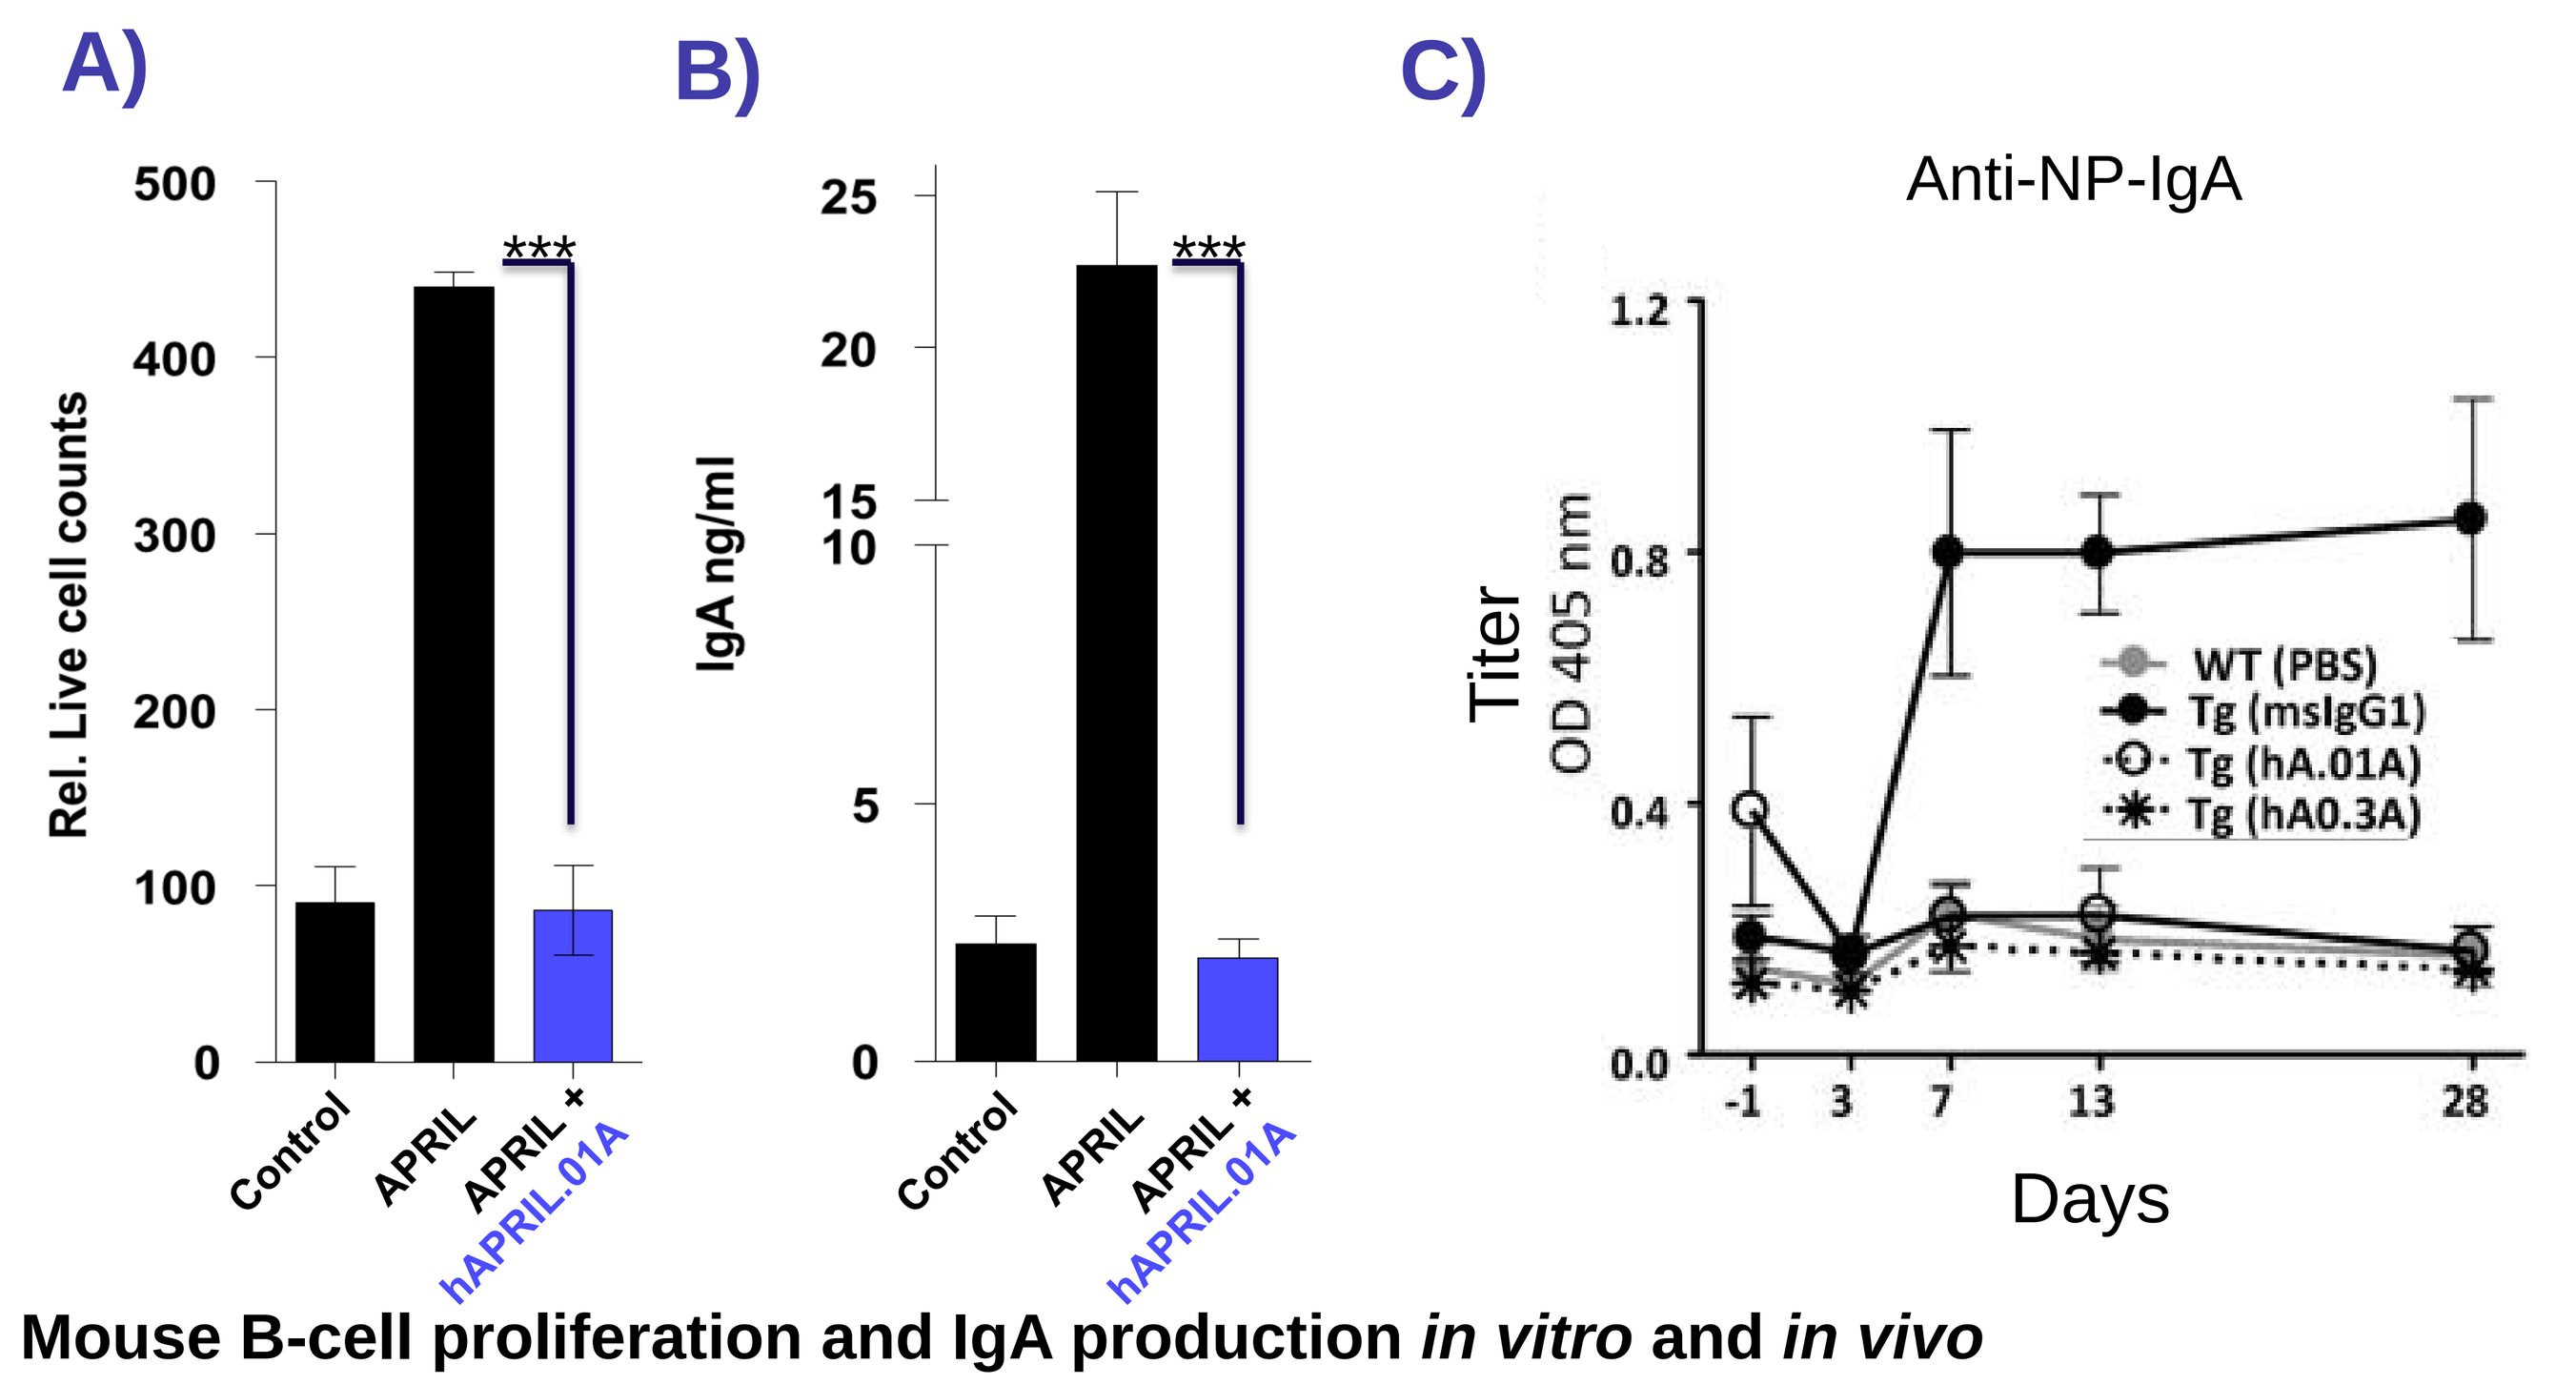
A)
B)
C)
Rel. Live cell counts
IgA ng/ml
Anti-NP-IgA
Titer
OD 405 nm
WT (PBS)
Tg (msIgG1)
Tg (hA.01A)
Tg (hA0.3A)
Days
Control
APRIL
APRIL + hAPRIL.01A

Mouse B-cell proliferation and IgA production *in vitro* and *in vivo*
Mouse B-cells were stimulated with recombinant FLAG-APRIL, and proliferation and IgA secretion was determined after 6 days of incubation. The mouse parental anti-APRIL antibody hAPRIL.01A inhibited APRIL-mediated B-cell proliferation (A) and IgA production (B). Human APRIL transgenic (Tg) or wild type (WT) mice were immunized with NP-Ficoll and humoral immune response was determined by detection of IgM, IgG and IgA antibody titers against NP-Ficoll antigen (C). Mice were treated with PBS, hAPRIL.01A (hA.01A), hAPRIL.03A (hA0.03A) or mouse IgG1 isotype-matched control. (A) Each sample was analyzed in triplicate. ***P < .001, 1-way ANOVA; error bars = SD. (Guadagnoli et al. Blood 2011)

Fig.4 BION-1301 Single Dose Pharmacokinetics

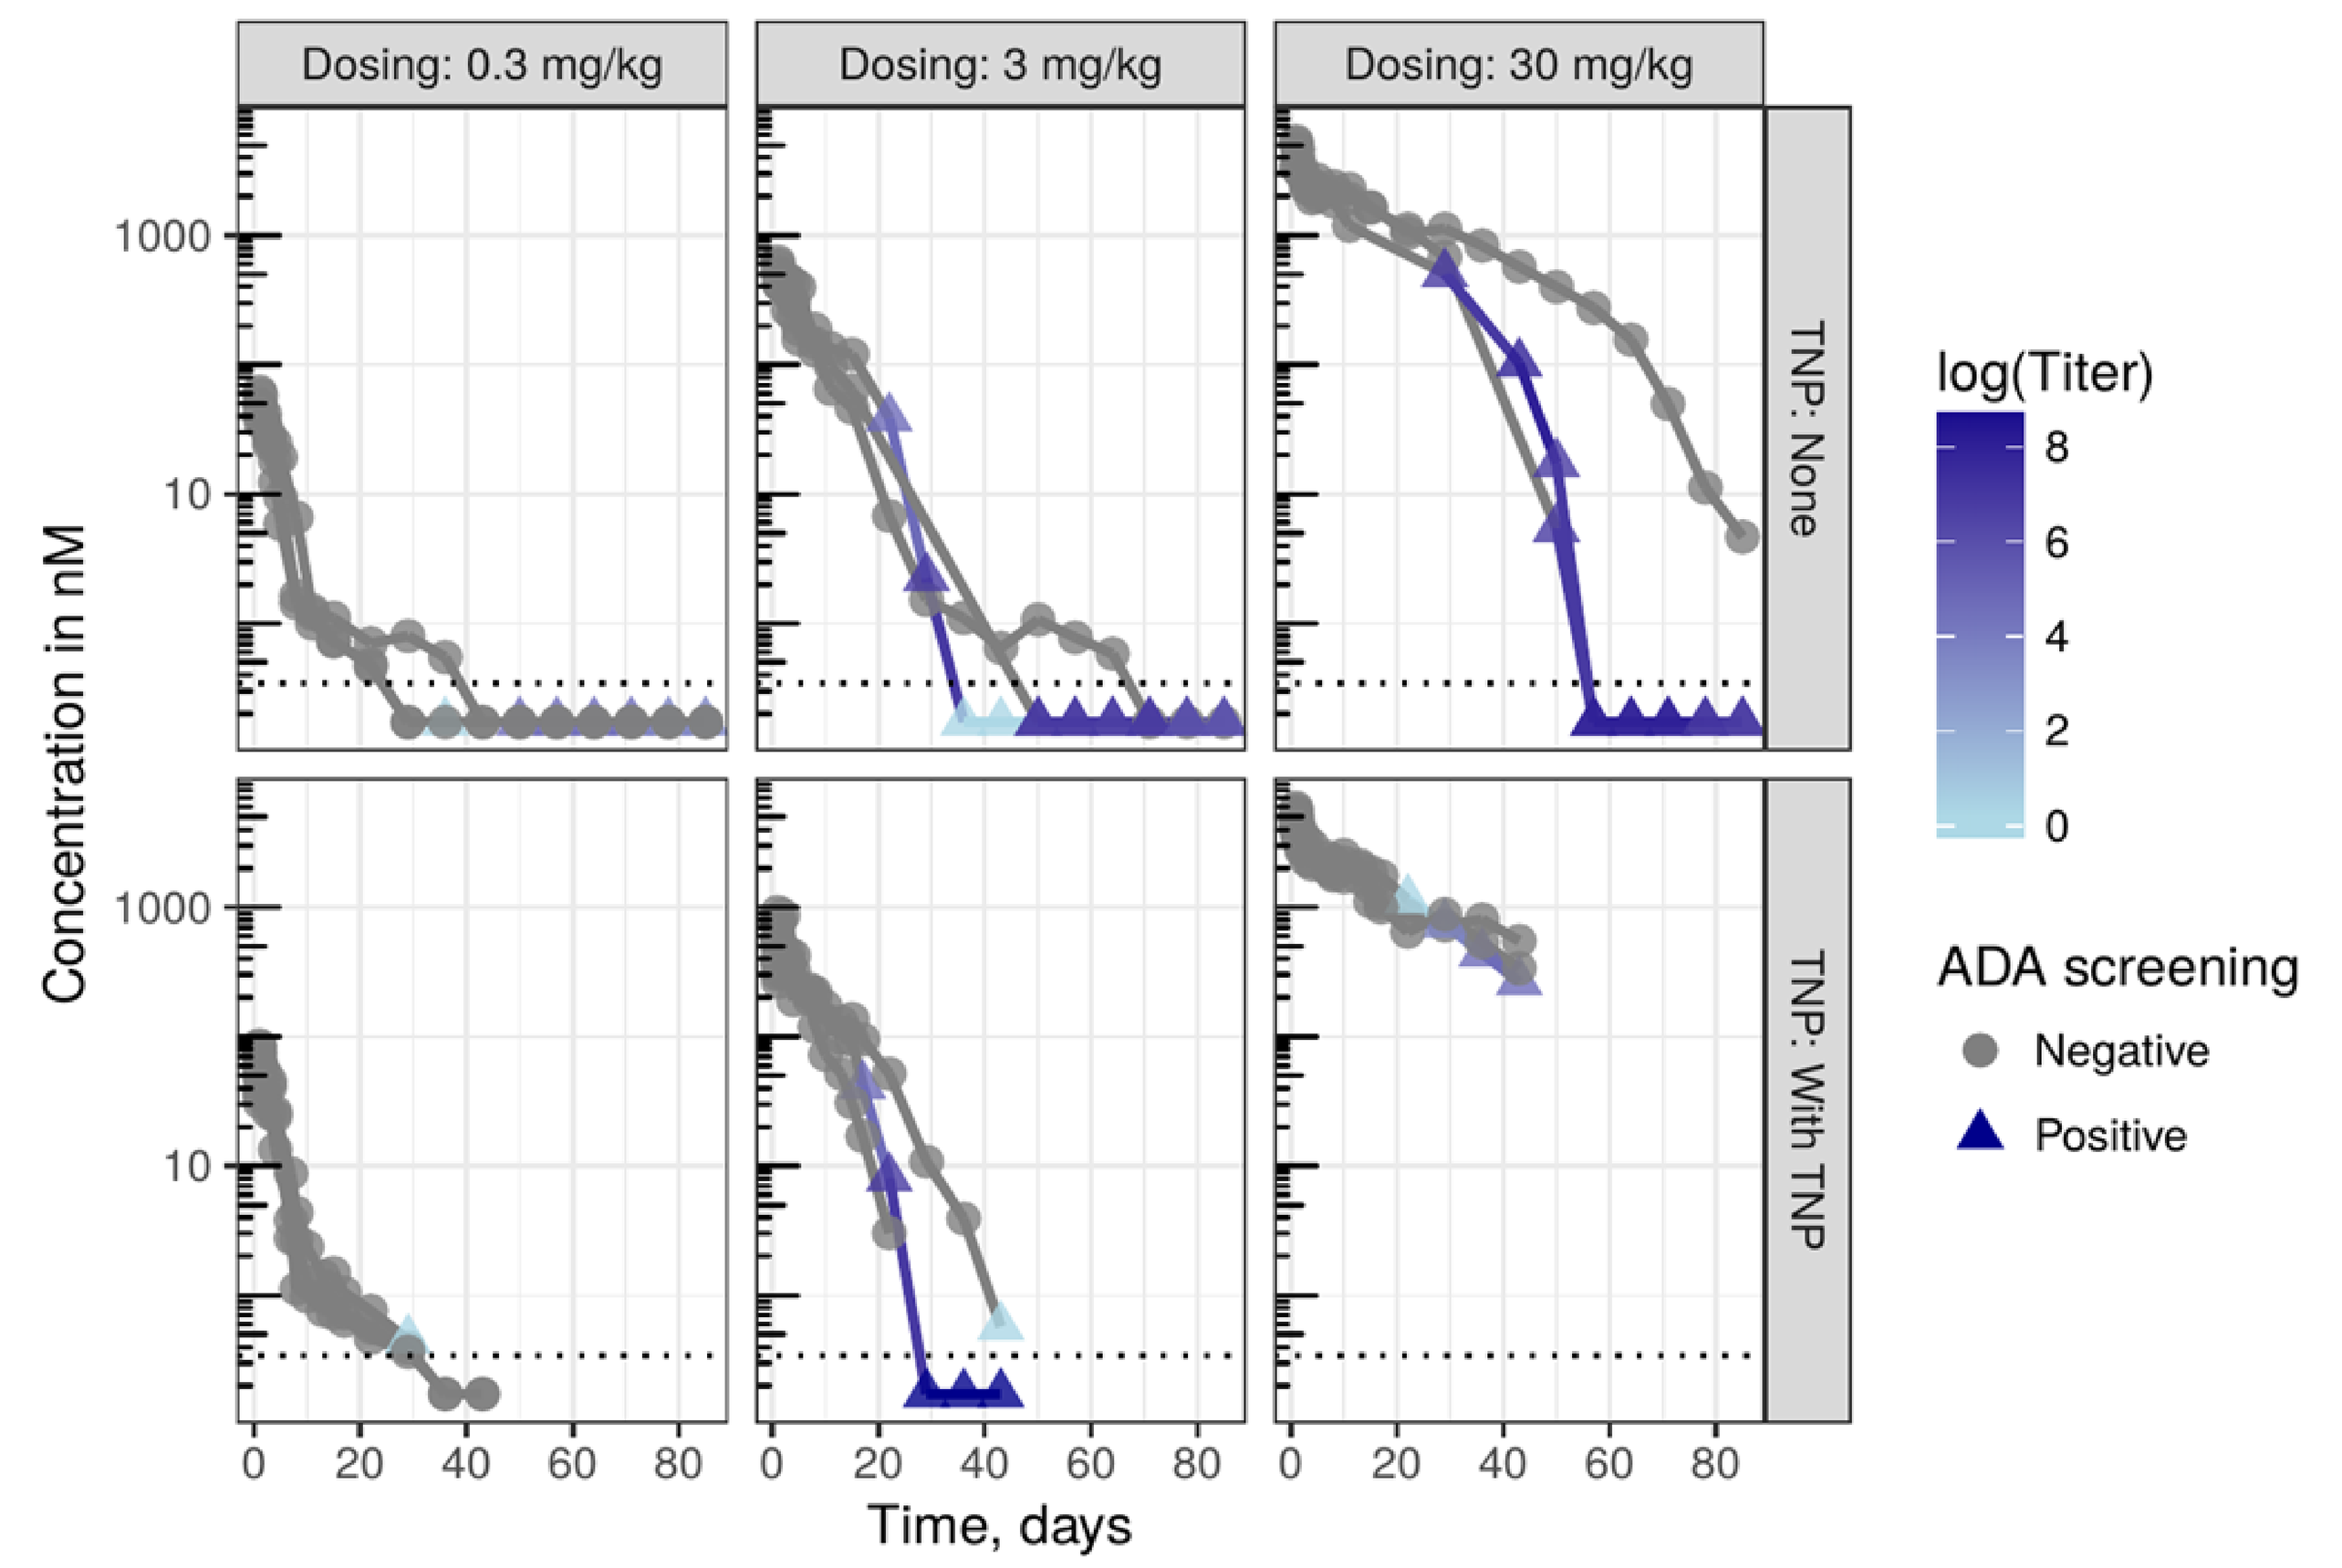
Dosing: 0.3 mg/kg
Dosing: 3 mg/kg
Dosing: 30 mg/kg
Concentration in nM
Time, days
TNP: None
TNP: With TNP
log(Titer)
ADA screening
Negative
Positive

Pharmacokinetics (PK) and anti-drug antibodies (ADA) following a single IV dose administration of BION-1301 to cynomolgus monkey
Serum BION-1301 concentration and anti-drug antibodies (ADA) were quantified by ELISA. PK of BION-1301 was not affected by TNP treatment and was generally characterized by a low clearance and limited distribution, as typical for antibodies. Light blue indicates that ADA was not confirmed and dark blue indicates a high ADA titer. A clear impact of ADA on PK was established in 2 out of 3 animals in the 30 mg/kg dose group. The terminal half life in NHP was estimated to be 7-13 days.

Fig.5 BION1301 parental antibody hAPRIL.01A Inhibits APRIL-Dependent Human Plasma-Blast/Cell Survival and IgA Production

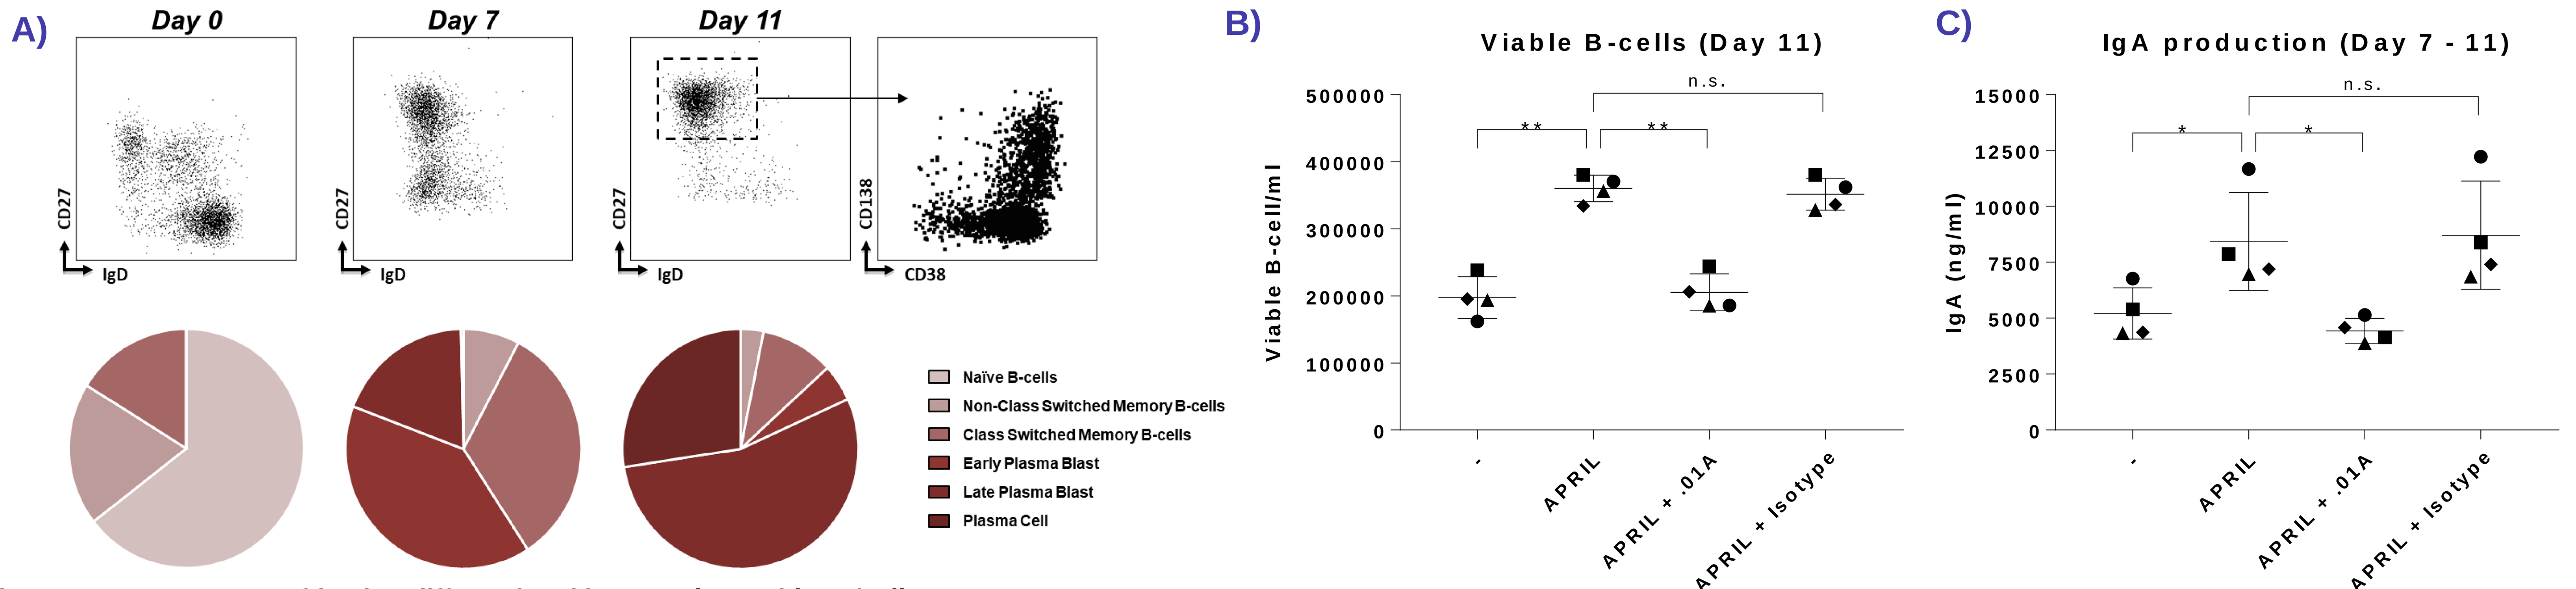
A)
Day 0
Day 7
Day 11
CD27
IgD
CD138
CD38
Naïve B-cells
Non-Class Switched Memory B-cells
Class Switched Memory B-cells
Early Plasma Blast
Late Plasma Blast
Plasma Cell
B)
Viable B-cells (Day 11)
Viable B-cell/ml
C)
IgA production (Day 7 - 11)
IgA (ng/ml)
-
APRIL
APRIL + .01A
APRIL + Isotype
n.s.

hAPRIL.01A treatment of *in vitro* differentiated human plasma-blasts/cells
Peripheral CD20+ B cells isolated from four healthy donors were differentiated to plasma-blasts/cells. At day 0, 7 and 11 viable B cells were assessed for B-cell differentiation markers (representative donor is shown (A)). From day 7-11, cells were cultured in the presence or absence of 100ng/mL human APRIL, APRIL + hAPRIL.01A (.01A) or APRIL + Isotype control mAb. On day 11, cells and supernatant were retrieved and the effect of anti-APRIL.01A on plasma-blast and plasma cell survival (B) and IgA production was assessed (C). Each symbol represents one donor. The line with error bars indicate the mean with SD. Asterisk indicate P-value (*:P<0.05; **:P<0.01; n.s.: not significant) for differences among condition by paired student T-test.

Fig.6 BION-1301 Suppresses Induced Hapten-Specific IgA as well as Total IgA, IgG and IgM in a Dose-Dependent Fashion

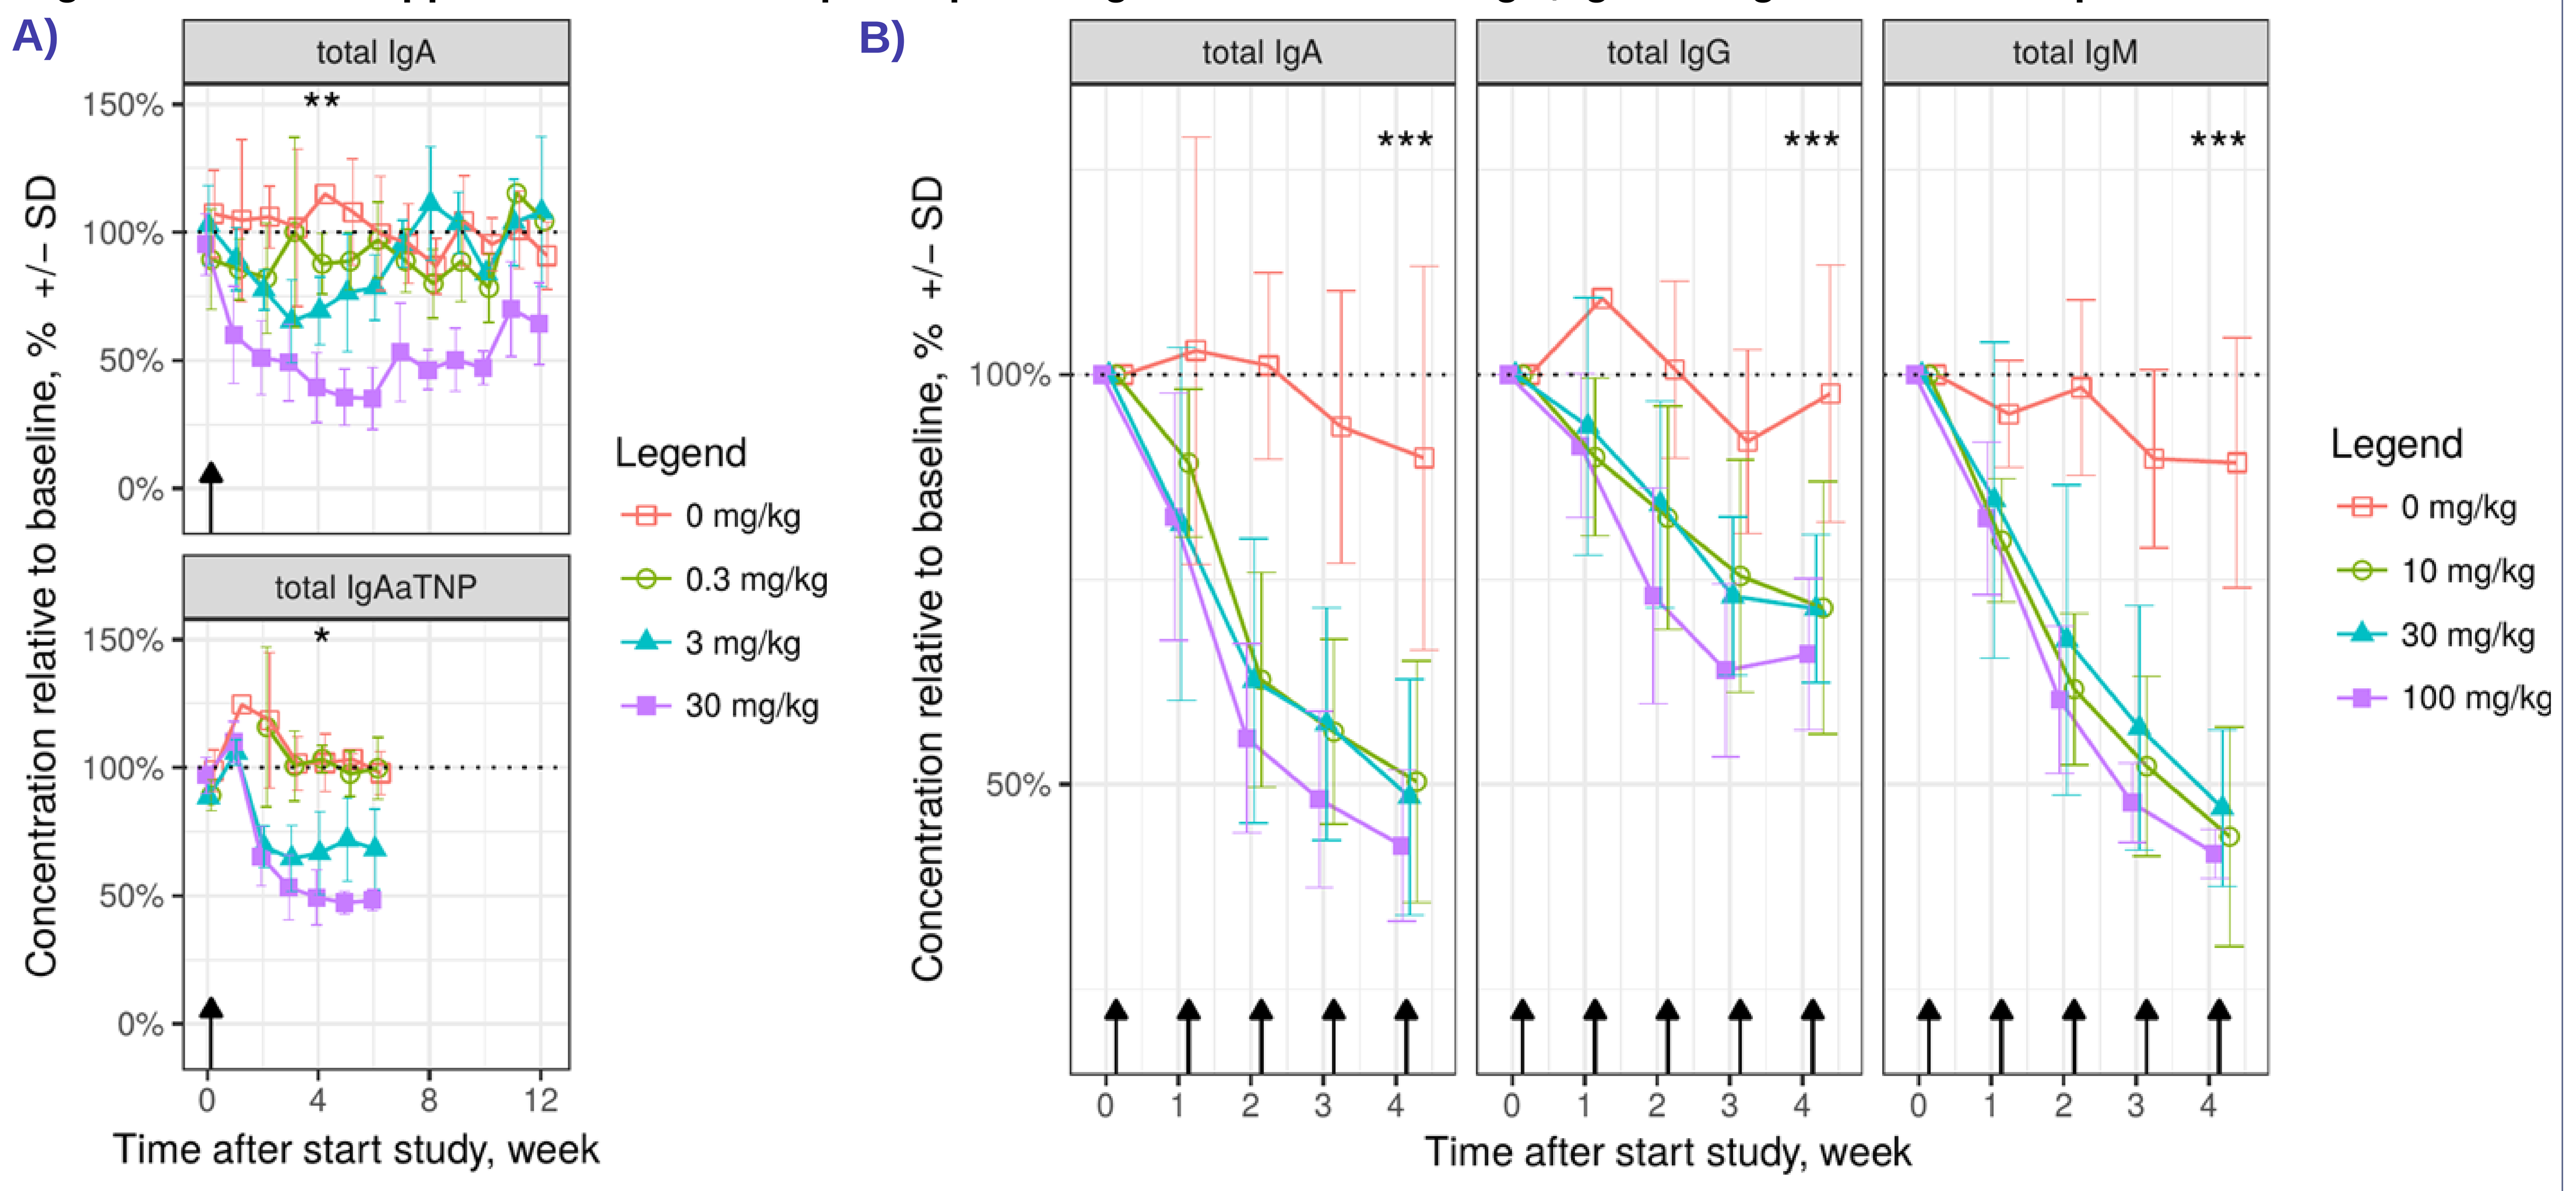
A)
total IgA
total IgAaTNP
Concentration relative to baseline, % +/− SD
Time after start study, week
Legend
0 mg/kg
0.3 mg/kg
3 mg/kg
30 mg/kg
B)
total IgA
total IgG
total IgM
Legend
0 mg/kg
10 mg/kg
30 mg/kg
100 mg/kg

Total and anti-TNP IgA level in single dose PK/PD study in NHP (A)
Total and anti-TNP (2,4,6-Trinitrophenyl) specific immunoglobulin type A (IgA) levels were determined after a single intravenous dose of BION-1301 in cynomolgus monkey. The upper panel contains immunoglobulin levels in BION-1301 only treated NHP, whereas the lower panel depicts anti-TNP specific levels. Arrows denote time of dosing and asterisks indicate P-value (*: P<0.05; **: P<0.01) for differences among treatment groups by non-parametric Kruskal-Wallis tests. Study design is shown in Figure 3A.

Total IgA, IgG and IgM level in multiple dose study in NHP (B)
Total Immunoglobulin levels were quantified during 4-week repeated intravenous dosing of BION-1301. Arrows denote time of dosing and asterisks, if present, indicate P-value (***: P<0.001) for differences among treatment groups by non-parametric Kruskal-Wallis tests. Study design is shown in Figure 3B.

CONCLUSION

We developed a novel humanized high-affinity anti-APRIL antibody BION-1301. Its epitope was mapped to the BCMA and TACI binding site conferring full blocking capacity to BION-1301. The mouse anti-human parental antibody hAPRIL.01A inhibited APRIL-dependent B-cell proliferation and IgA production *in vitro* and *in vivo*. BION-1301 suppresses the T-cell independent (TI) B-cell response in NHP, confirming its suppressive activity in the TI mouse model. In human B-cell cultures, hAPRIL.01A reduced the number of IgA secreting plasma-blasts/ cells. Exposure to BION-1301 induced a significantly reduced IgA levels vs baseline of -50±14, -52±14, -59±7 % at 10, 30 and 100 mg/kg respectively in NHP. In a 4-week repeat dose NHP study BION-1301 showed a favorable safety profile. In summary, we report the *in vivo* and *in vitro* mechanism of action of a novel humanized APRIL neutralizing antibody BION-1301 modulating IgA, supporting its development for the treatment of IgAN.

Reference:
Guadagnoli M, Kimberley FC, Phan U, Cameron K, Vink PM, Rodermond H, Eldering E, Kater AP, van Eenennaam H, Medema JP. Development and characterization of APRIL antagonistic monoclonal antibodies for treatment of B-cell lymphomas. *Blood*. 2011 Jun 23;117(25):6856-65.

Presented at 2018 American Society of Nephrology, October 23-28, 2018, San Diego, CA